A Phase 1 Dose-Escalation Study of the Novel Cell Cycle Active Agent SNS-595 in Advanced Leukemias

J.Lancet[1], J.Karp[2], M.Suster[3], U.Hoch[3], J.Silverman[3], G. Michelson[3], F.Giles[4]

[1]H. Lee Moffitt Cancer Center and Research Institute, [2]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, [3]Sunesis Pharmaceuticals, Inc., [4]MD Anderson Cancer Center

UPDATED ABSTRACT

SNS-595 is a novel, cell cycle-active cytotoxic naphthyridine analog that induces G2 arrest in a variety of preclinical tumor models. We initiated an escalating-dose phase 1 trial of SNS-595 Injection, administered as a weekly x 3 (arm A) or twice weekly x 2 bolus (arm B), in patients with advanced or refractory acute leukemias.

Objectives: The primary objectives were to: 1) establish safety, tolerability, and MTD of SNS-595 given on each schedule, 2) characterize pharmacokinetics (PK) of SNS-595 when given on these schedules. Secondary objectives were: 1) assessment of clinical activity, 2) exploration of potential biomarkers.

Methods: SNS-595 Injection was administered as a slow IV push on days 1, 8, 15 (arm A) or days 1, 4, 8, 11 (arm B). Minimum cycle length was 42 days (arm A) and 39 days (arm B). Additional cycles were permitted if patients achieved stable disease or better. The starting dose was 18 mg/m^2/d on arm A, and 9 mg/m^2/d on arm B and escalated by cohort using a modified Fibonacci schema. PK analyses for SNS-595 were performed on plasma samples collected during cycle 1. Pretreatment peripheral blood and bone marrow aspirate samples were collected for exploratory analyses of the level and functional activity of the DNA damage repair proteins DNA-PK and MSH2.

Results: To date, 31 patients have been enrolled and are evaluable in the live database, including 17 patients assigned to arm A and 14 assigned to arm B, 17 males and 14 females with a median age of 66 years. Diagnoses included AML (29 patients) and ALL (2 patients). All patients had disease refractory to and/or relapsed from prior therapy (median 3 prior regimens (range 1-5)). Dose escalation has proceeded to 50 mg/m^2/d (arm A) and 25 mg/m^2/d (arm B). One dose-limiting toxicity has been observed to date. Non-dose limiting toxicities included nausea/vomiting, diarrhea, and mucositis. Grade 4 neutropenic fever was observed in only one patient. Plasma exposures at the first three or four dose levels in each arm increased linearly, resulting in AUCs of 4.3 to 25 μg•hr/mL for 9-50 mg/m^2 doses. CL, Vss, and terminal half-lives were similar to those reported previously in solid tumor patients, and averaged ~2 L/hr/m^2, 63 L/m^2, and 24 hr, respectively. No patients have achieved complete response to date, although 3 patients (out of the 5 in the 50 mg/m^2/d cohort) experienced clinically significant reductions in bone marrow blasts to $\leq$ 5% during cycle 1.

Conclusion: SNS-595 appears to be well-tolerated in patients with advanced leukemias, with preliminary and promising signs of clinical activity as measured by decreases in leukemic blasts. Evidence of bone marrow ablation is accumulating; patient accrual and dose-escalation are ongoing.

BACKGROUND

SNS 595 is a novel naphthyridine analog, a class of compounds not previously used for cancer treatment, and is a cell cycle-active agent that acts through DNA-PK signaling in the S phase to induce apoptosis and a G2-phase arrest of the cell cycle. Neutropenia was the dose limiting toxicity in two phase 1 studies (Proc ASCO 2006). Preclinically, SNS-595 has been shown to reduce bone marrow cellularity and circulating neutrophils in mice in a dose-dependent fashion (Proc AACR 2006) and is synergistic with cytarabine in models (Proc ASH 2006). Preliminary results are presented of a clinical study in acute leukemia to explore the safety, tolerability and preliminary clinical activity of SNS-595 in acute hematologic malignancies.

STUDY OBJECTIVES

- Determine the safety and tolerability of escalating doses of SNS-595 Injection administered to patients assigned to one of two dosing schedules
 - Schedule A: Once-weekly IV administration of SNS-595 Injection (Days 1, 8, and 15) for 3 doses per cycle
 - Schedule B: Twice-weekly IV administration of SNS-595 Injection (Days 1, 4, 8, and 11) for 4 doses per cycle
- Assess the PK profile of SNS-595 in patients with advanced hematologic malignancies
- Define a recommended dose regimen for future phase 2 studies
- Obtain preliminary assessments of antileukemia activity

METHODS AND SCHEMA

- DLT Definition:
 - NCI CTCAE Grade 4 hematologic event(s) of neutropenia or thrombocytopenia occurring thru Cycle 1 Day 29 that are assessed as clinically significant and related to study drug and that persist in the absence of viable leukemia beyond 8 weeks after the Cycle 1 Day 1 dose
 - $\geq$ NCI CTCAE Grade 3 nonhematologic event(s) occurring thru Cycle 1 Day 29 that are assessed as clinically significant and related to study drug, regardless of duration.
- Drug was administered at a starting dose of 18 mg/m^2 IV weekly for 3 weeks (Schedule A) or 9 mg/m^2 IV given twice weekly for 2 weeks (Schedule B). Dose escalation followed a modified Fibonacci schema.
- Major Entry Criteria: Relapsed or refractory leukemia; $\leq$ 3 induction/re-induction regimens for either AML or ALL; adequate renal and hepatic function

PATIENTS

Table 1: Patient Demographics

	qw x3	biw x4	total
n (# treated)	17	14	31
Sex			
Male	10 (59%)	7 (50%)	17 (55%)
Female	7 (41%)	7 (50%)	14 (45%)
Ethnic Background			
Black	2 (12%)	1 (7%)	3 (10%)
Hispanic	2 (12%)	2 (14%)	4 (13%)
White	13 (77%)	11 (79%)	24 (77%)
Age (yrs)			
Mean	59.2	62.7	60.8
Median	66	68	66
Range	21-77	28-85	21-85

Table 2: Baseline Characteristics

	qw x3	biw x4	total
n (# treated)	17	14	31
Diagnosis			
AML	16 (94%)	13 (93%)	29 (94%)
ALL	1 (6%)	1 (7%)	2 (7%)
Disease Status			
Relapsed	2 (12%)	4 (29%)	6 (19%)
Refractory	8 (47%)	6 (43%)	14 (45%)
Relapsed and Refractory	6 (35%)	4 (29%)	10 (32%)
ECOG Performance			
0	5 (29%)	5 (36%)	10 (32%)
1	11 (65%)	8 (57%)	19 (61%)
2	1 (6%)	1 (7%)	2 (7%)
Median # Previous Leukemia Therapies	3	3	3

SAFETY DATA

SNS-595 is well-tolerated

- One dose-limiting toxicity has been observed to date: prolonged myelosuppression
- Grade 4 neutropenic fever was observed in one patient and was attributed to study drug

Table 3: Study drug-related AEs (all CTCAE Grades)

	Schedule A (qw x3)				Schedule B (biw x4)				
assigned dosage (mg/m^2)	18	27	38	50	9	14	19	25	total
# Patients Treated	3	6	3	5	4	4	3	3	31
# Patients Reporting AEs	**2**	**1**	**0**	**0**	**3**	**1**	**1**	**0**	**8**
Nausea	1	0	0	0	3	1	0	0	5
Vomiting	0	0	0	0	2	1	0	0	3
Diarrhoea	0	0	0	0	1	0	0	0	2
Diarrhoea					1				
Deep vein thrombosis	0	1	0	0	0	0	0	0	1
Thrombophlebitis superficial	1	0	0	0	0	0	0	0	1
Myelosuppression	0	0	0	1	0	0	0	0	1
Alopecia	0	0	0	0	0	0	1	0	1
Anorexia	0	0	0	0	1	0	0	0	1
Mucosal inflammation	0	0	0	0	1	0	0	0	1
Febrile neutropenia	0	0	0	0	1	0	0	0	1

Pink cells signify CTCAE Grade 3 or 4: Rose for Grade 3, Fuchsia for Grade 4

PHARMACOKINETICS

SNS-595 shows predictable and highly reproducible pharmacokinetics

- Dose dependent increase in exposure and no change in CL or Vss with dose and schedule
- No accumulation or change in pharmacokinetic parameters after repeat dosing

Table 4: Pharmacokinetics (mean ± SD)

Dose (mg/m^2)	Schedule	Day	n	T ½ (hr)	CL (L/hr/m^2)	Vss (L/m^2)	AUC (ughr/ml)
9*	B	1	4	24 ± 6	2.3 ± 0.7	65 ± 2	4.3 ± 1.3
		11	4	26 ± 11	2.0 ± 1.2	59 ± 24	6.7 ± 5.4
13.5*	B	1	4	25 ± 7	2.5 ± 0.7	75 ± 20	5.7 ± 1.7
		11	3	22 ± 8	2.4 ± 0.7	61± 30	5.9 ± 1.9
18	A	1	3	24 ± 4	2.3 ± 0.5	72 ± 21	8.0 ± 1.4
19*	B	1	3	28 ± 1	2.2 ± 0.5	81± 17	8.8 ± 2.0
		11	3	17 ± 6	2.4 ± 0.4	52 ± 13	8.1± 1.4
27	A	1	5	24 ± 12	1.6 ± 0.5	48 ± 22	17.8 ± 5.1
38	A	1	3	24 ± 8	2.7 ± 0.8	62 ± 4	15.0 ± 5.1
50	A	1	2	28	2	57	25

** Similar PK after Days 4, and 8 administration*

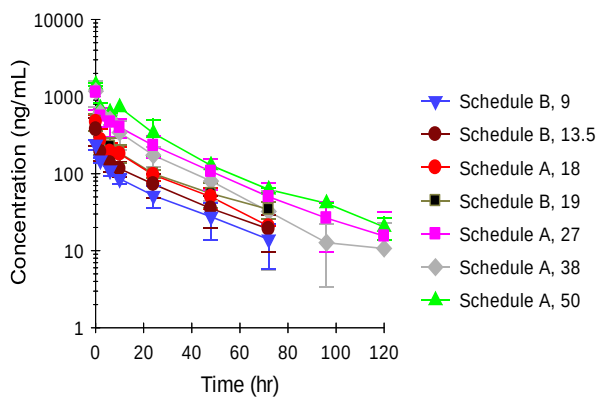
10000
1000
100
10
1
Concentration (ng/mL)
0
20
40
60
80
100
120
Time (hr)
Schedule B, 9
Schedule B, 13.5
Schedule A, 18
Schedule B, 19
Schedule A, 27
Schedule A, 38
Schedule A, 50

Figure 1: Plasma concentration-time profile for schedule A (qw x3) and B (biw x4)

EVIDENCE OF ACTIVITY

SNS-595 reduces bone marrow blasts in 3 out of the 5 relapsed/refractory poor prognosis AML patients dosed at 50 mg/m^2, a level expected to be near the MTD based on preclinical models

- 75 year old African-American female with relapsed AML showed bone marrow blast decrease from 70-80% to <5% after initial therapy with SNS-595 *(previously failed treatment with tipifarnib and etoposide)*
- 78 year old African-American male with relapsed/refractory secondary AML (from MDS) and poor risk cytogenetics showed bone marrow blast decrease from 80% to 2% after initial therapy with SNS-595 *(previously failed treatment with "7 +3", HiDAC, gemtuzumab ozogamicin, and azacitidine)*
- 66 year old Caucasian male with primary refractory AML and normal cytogenetics showed bone marrow blast decrease from 31% to 5% after initial treatment with SNS-595 *(previously failed treatment with"7 + 3" x 2, and 2-CDA/Ara-C)*

CONCLUSIONS AND FUTURE DIRECTIONS

SNS-595 is a novel cell cycle inhibitor with a unique mechanism of action

- ✓ **Demonstrates evidence of clinical activity in relapsed/refractory AML patients and was well tolerated**
- ✓ **Predictable and highly reproducible pharmacokinetics**
- ✓ **MTD not yet reached, dose escalation continues**
- ✓ **Phase 1b combination, SNS-595 and cytarabine, planned for 2007**
- ✓ **Phase 2 trials in ovarian cancer, NSCLC, and SCLC are in progress**

Disclosures: JL, JK, and FG receive research funding from Sunesis. MS, UH, JS, and GM are employees of Sunesis Pharmaceuticals Inc. and may possess corresponding shares and/or options.